In Treatment-Naive Patients with Relapsing-Remitting Multiple Sclerosis (RRMS), Initiating Natalizumab Earlier Is Associated with Greater Disability Improvement Than Delaying Treatment: Real-World Results from the TYSABRI® Observational Program (TOP)

P350

Spelman T,[1] Butzkueven H,[1,2] Kappos L,[3] Trojano M,[4] Wiendl H,[5] Chen Y,[6] Campbell N,[7] Licata S,[7] Ho PR,[7] on behalf of the TOP Investigators

[1]University of Melbourne, Melbourne, Australia; [2]Monash University, Box Hill, Australia; [3]University Hospital and University of Basel, Basel, Switzerland; [4]University of Bari, Bari, Italy; [5]University of Münster, Münster, Germany; [6]Biogen, Cambridge, MA, during completion of work; [7]Biogen, Cambridge, MA

Conclusions

- In TOP RRMS patients who were treatment naive when starting natalizumab, annualized relapse rates were significantly reduced from baseline and rates of Expanded Disability Status Scale (EDSS) worsening remained low over 3 years of treatment.
 - The effectiveness of natalizumab treatment was observed regardless of baseline disease activity.
- Patients initiating natalizumab soon after multiple sclerosis (MS) symptom onset had a higher probability of on-treatment disability improvement (ie, confirmed EDSS improvement) than patients initiating later.
 - The potential to improve patient function may be an important outcome measure to consider in the benefit-risk assessment of high-efficacy MS therapies.
 - The association of earlier natalizumab treatment with an increased probability of disability improvement should also be considered when evaluating a patient's individualized benefit-risk.
- These results demonstrate that patients who initiate natalizumab treatment earlier in their disease course have better outcomes, which is consistent with previous analyses of patients in TOP.[1-3]
 - Disability early in the course of MS may reflect inflammation that can be mitigated by timely treatment with natalizumab, whereas later disability may be more resistant to recovery.

Introduction

- Natalizumab is an effective treatment for MS that has been shown to reliably reduce relapse rates and disability worsening in both clinical trials and real-world clinical practice.[3,4]
 - In particular, natalizumab treatment is associated with lower clinical measures of disease activity when initiated earlier in the disease course.[3,5]
- Post hoc analyses of clinical trials and data from real-world observational studies have demonstrated the association of natalizumab treatment with improvements in disability.[6-10]
- Additional real-world data confirming the relationship between natalizumab use early in the disease course and achievement of sustained disease control and improved function may help guide benefit-risk assessments during treatment decision-making.
- TOP is an ongoing 10-year observational study providing long-term data on RRMS patients initiating natalizumab in clinical practice settings.[3]

Objective

- To compare the real-world outcomes for treatment-naive patients starting natalizumab shortly after MS symptom onset with outcomes for patients initiating natalizumab later.

Methods

Patients

- Treatment-naive patients who enrolled in TOP and initiated natalizumab by May 1, 2016, were included in this analysis.
- Patients in TOP received 300 mg intravenous natalizumab every 4 weeks.
- Patient cohorts were determined according to RRMS disease duration, defined as time since symptom onset: ≤1 year (n=233), 1–5 years (defined as >1 and ≤5 years; n=179), and >5 years (n=144).

Analyses

- Annualized relapse rates in the year prior to natalizumab initiation and while on natalizumab were compared.
- Disability outcomes were assessed over 3 years of treatment. The number of patients in each cohort of treatment-naive patients with data for treatment beyond 3 years was limited.
- In this analysis, 1 year was defined as 12 months consisting of 4 weeks each.
- The Kaplan-Meier method was used to estimate the cumulative probability of confirmed EDSS worsening or improvement.
- Confirmed EDSS worsening was defined as a score increase of ≥1.5 from a baseline score of 0.0, an increase of ≥1.0 from a baseline score of 1.0–5.5, or an increase of ≥0.5 from a baseline score ≥6.0, confirmed 24 weeks later.
- Confirmed EDSS improvement was defined as a score decrease of ≥1.0 from a baseline score ≥2.0, confirmed 24 weeks later. Patients with a baseline EDSS score <2.0 were not included in the EDSS improvement analysis.
- Multivariate models were adjusted for age, relapses in the previous year, and baseline EDSS score.

Results

Patients

- A total of 556 patients who were treatment naive prior to natalizumab initiation were enrolled in TOP as of May 1, 2016.
- Treatment-naive patients with ≤1 year or 1–5 years since MS symptom onset were generally younger than treatment-naive patients with >5 years since onset. (Table 1)
- Compared with patients with >5 years since MS symptom onset, patients earlier in the disease course had more active disease at baseline as assessed by gadolinium-enhancing (Gd+) lesions and the number of relapses in the prior year.

Annualized Relapse Rates

- Natalizumab treatment significantly reduced annualized relapse rates compared with baseline in all 3 cohorts. (Figure 1).
 - The magnitude of reduction was consistent across all groups (92.6%–92.8%).

Change in Disability

- EDSS scores were significantly lower at year 3 than at baseline in the patient group with ≤1 year since MS symptom onset but not in the groups with longer disease duration. (Figure 2)
- Over 3 years of natalizumab treatment, the adjusted likelihood of EDSS worsening did not differ significantly between patients with ≤1 year since MS symptom onset (12.8%) and those with 1–5 years (23.2%; *P*=0.0584) or >5 years (13.4%; *P*=0.6419) since symptom onset. (Figure 3)
- The adjusted likelihood of EDSS improvement was greater for patients with ≤1 year since MS symptom onset (49.3%) than for those with 1–5 years (38.1%; *P*=0.0212) or >5 years (26.3%; *P*=0.0029) since symptom onset. (Figure 4)

Figure 1. Annualized relapse rates in the year prior to starting natalizumab and while on natalizumab

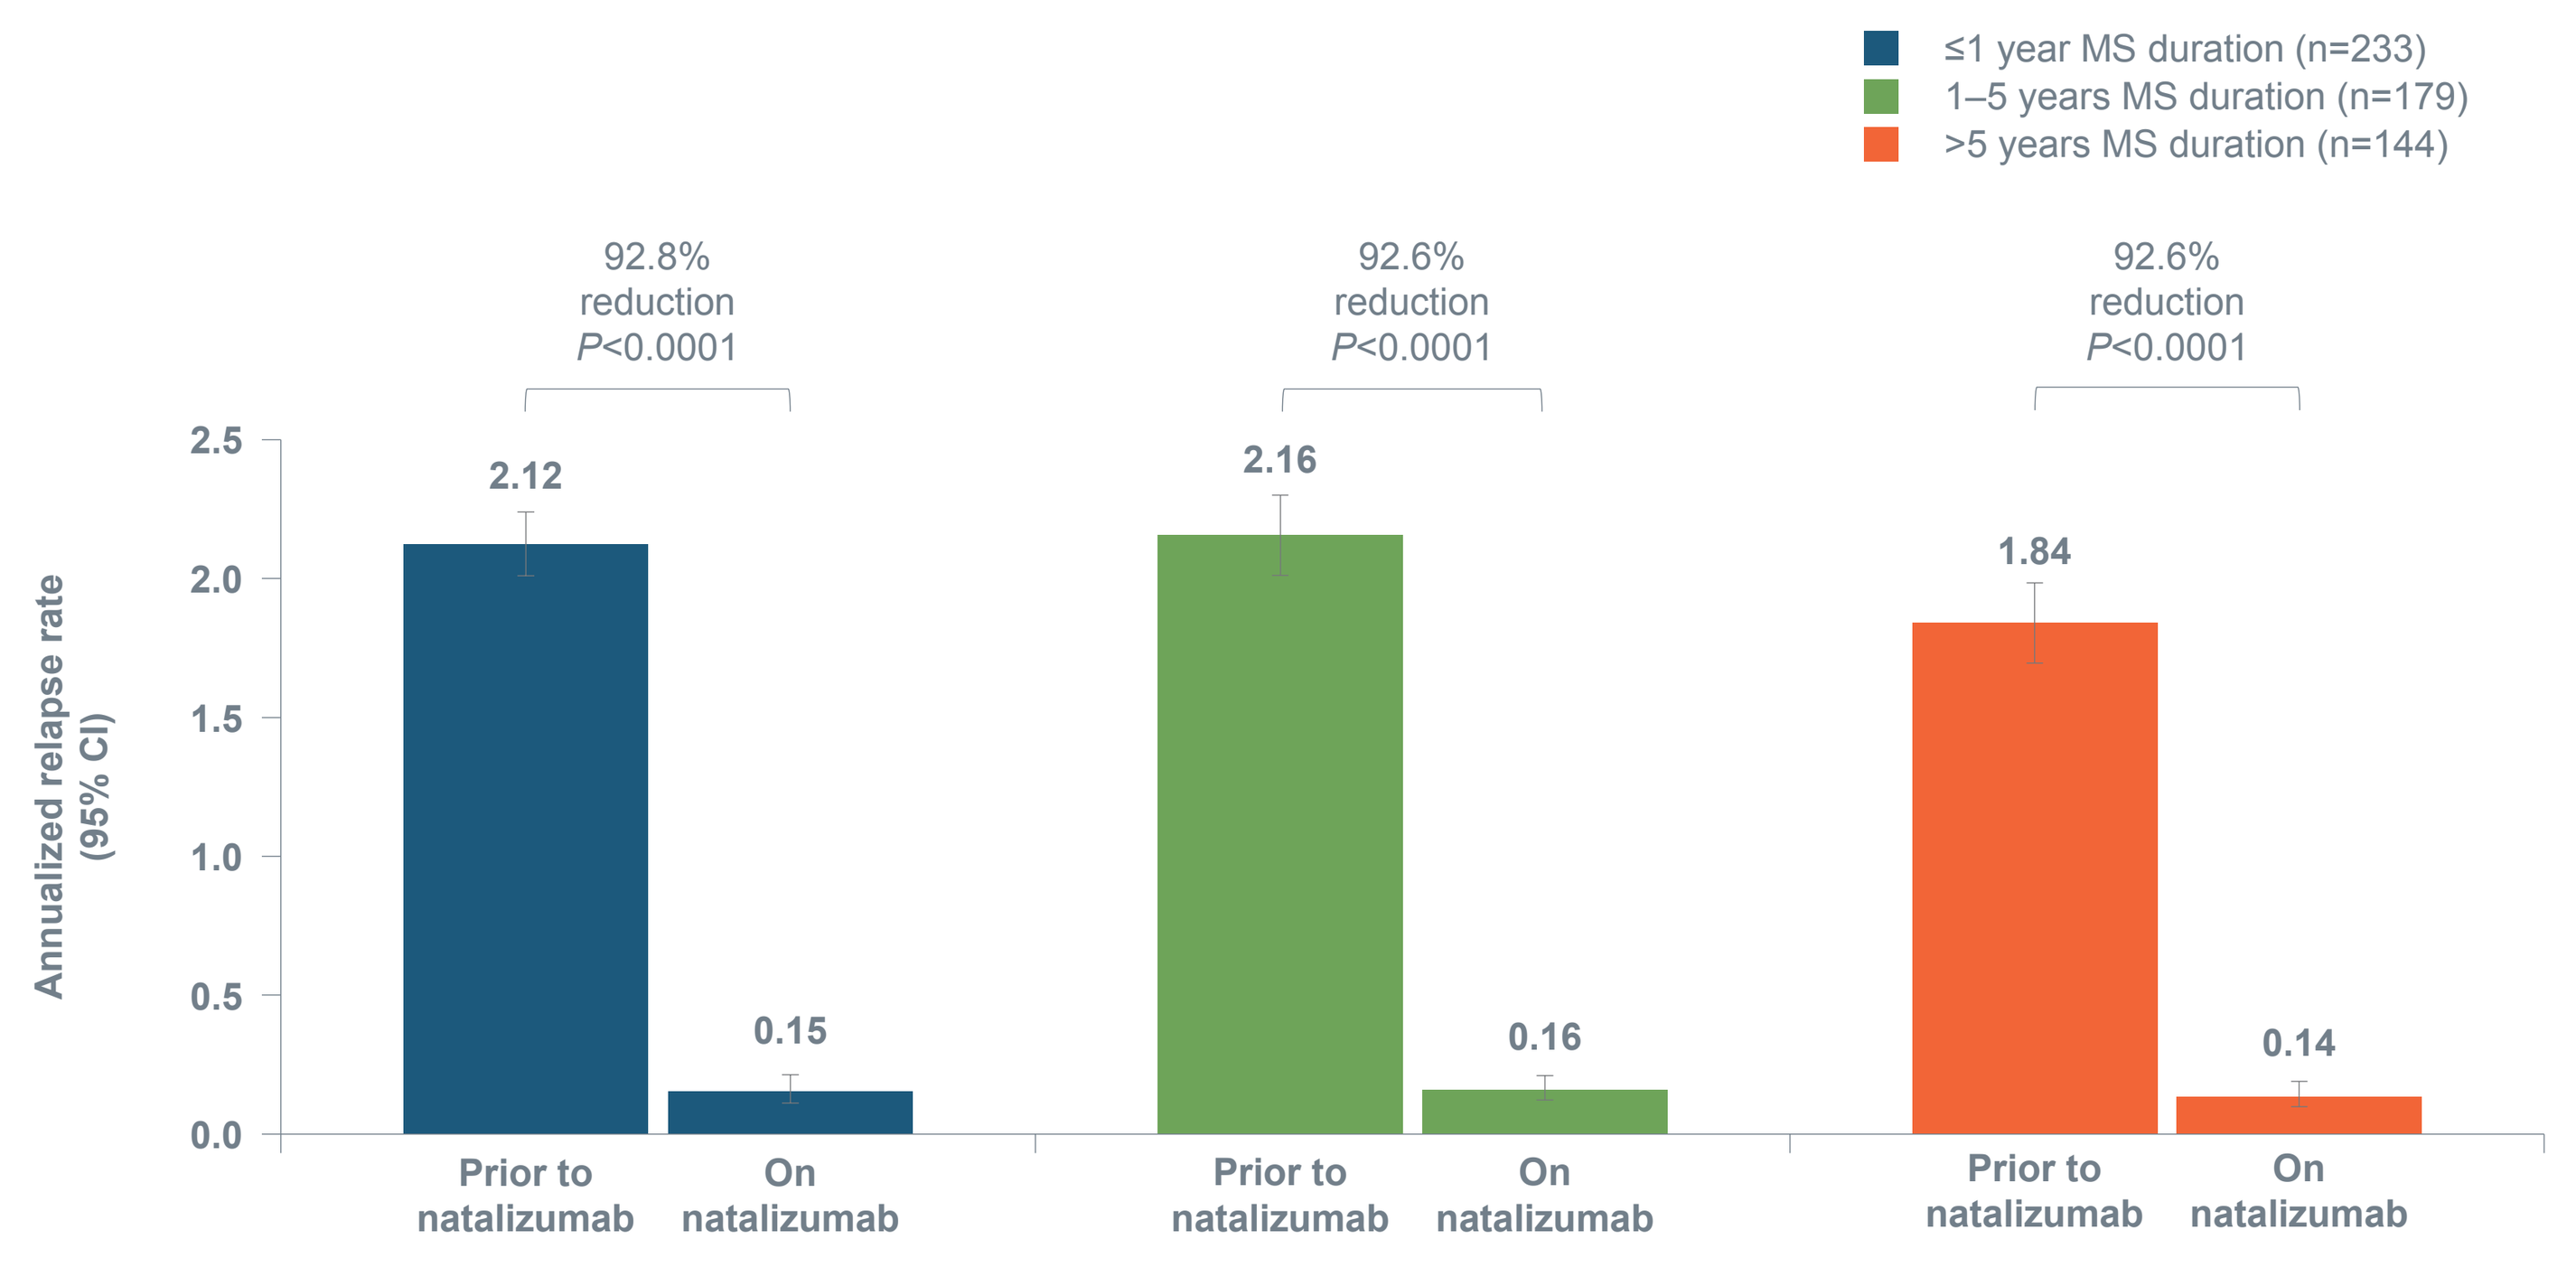
≤1 year MS duration (n=233)
1–5 years MS duration (n=179)
>5 years MS duration (n=144)
92.8% reduction P<0.0001
92.6% reduction P<0.0001
92.6% reduction P<0.0001
Annualized relapse rate (95% CI)
2.12
0.15
2.16
0.16
1.84
0.14
Prior to natalizumab
On natalizumab

CI=confidence interval.
P value is based on a Poisson model assessing within-group annualized relapse rate reduction from baseline.

Figure 2. EDSS score change from baseline at 3 years of natalizumab treatment

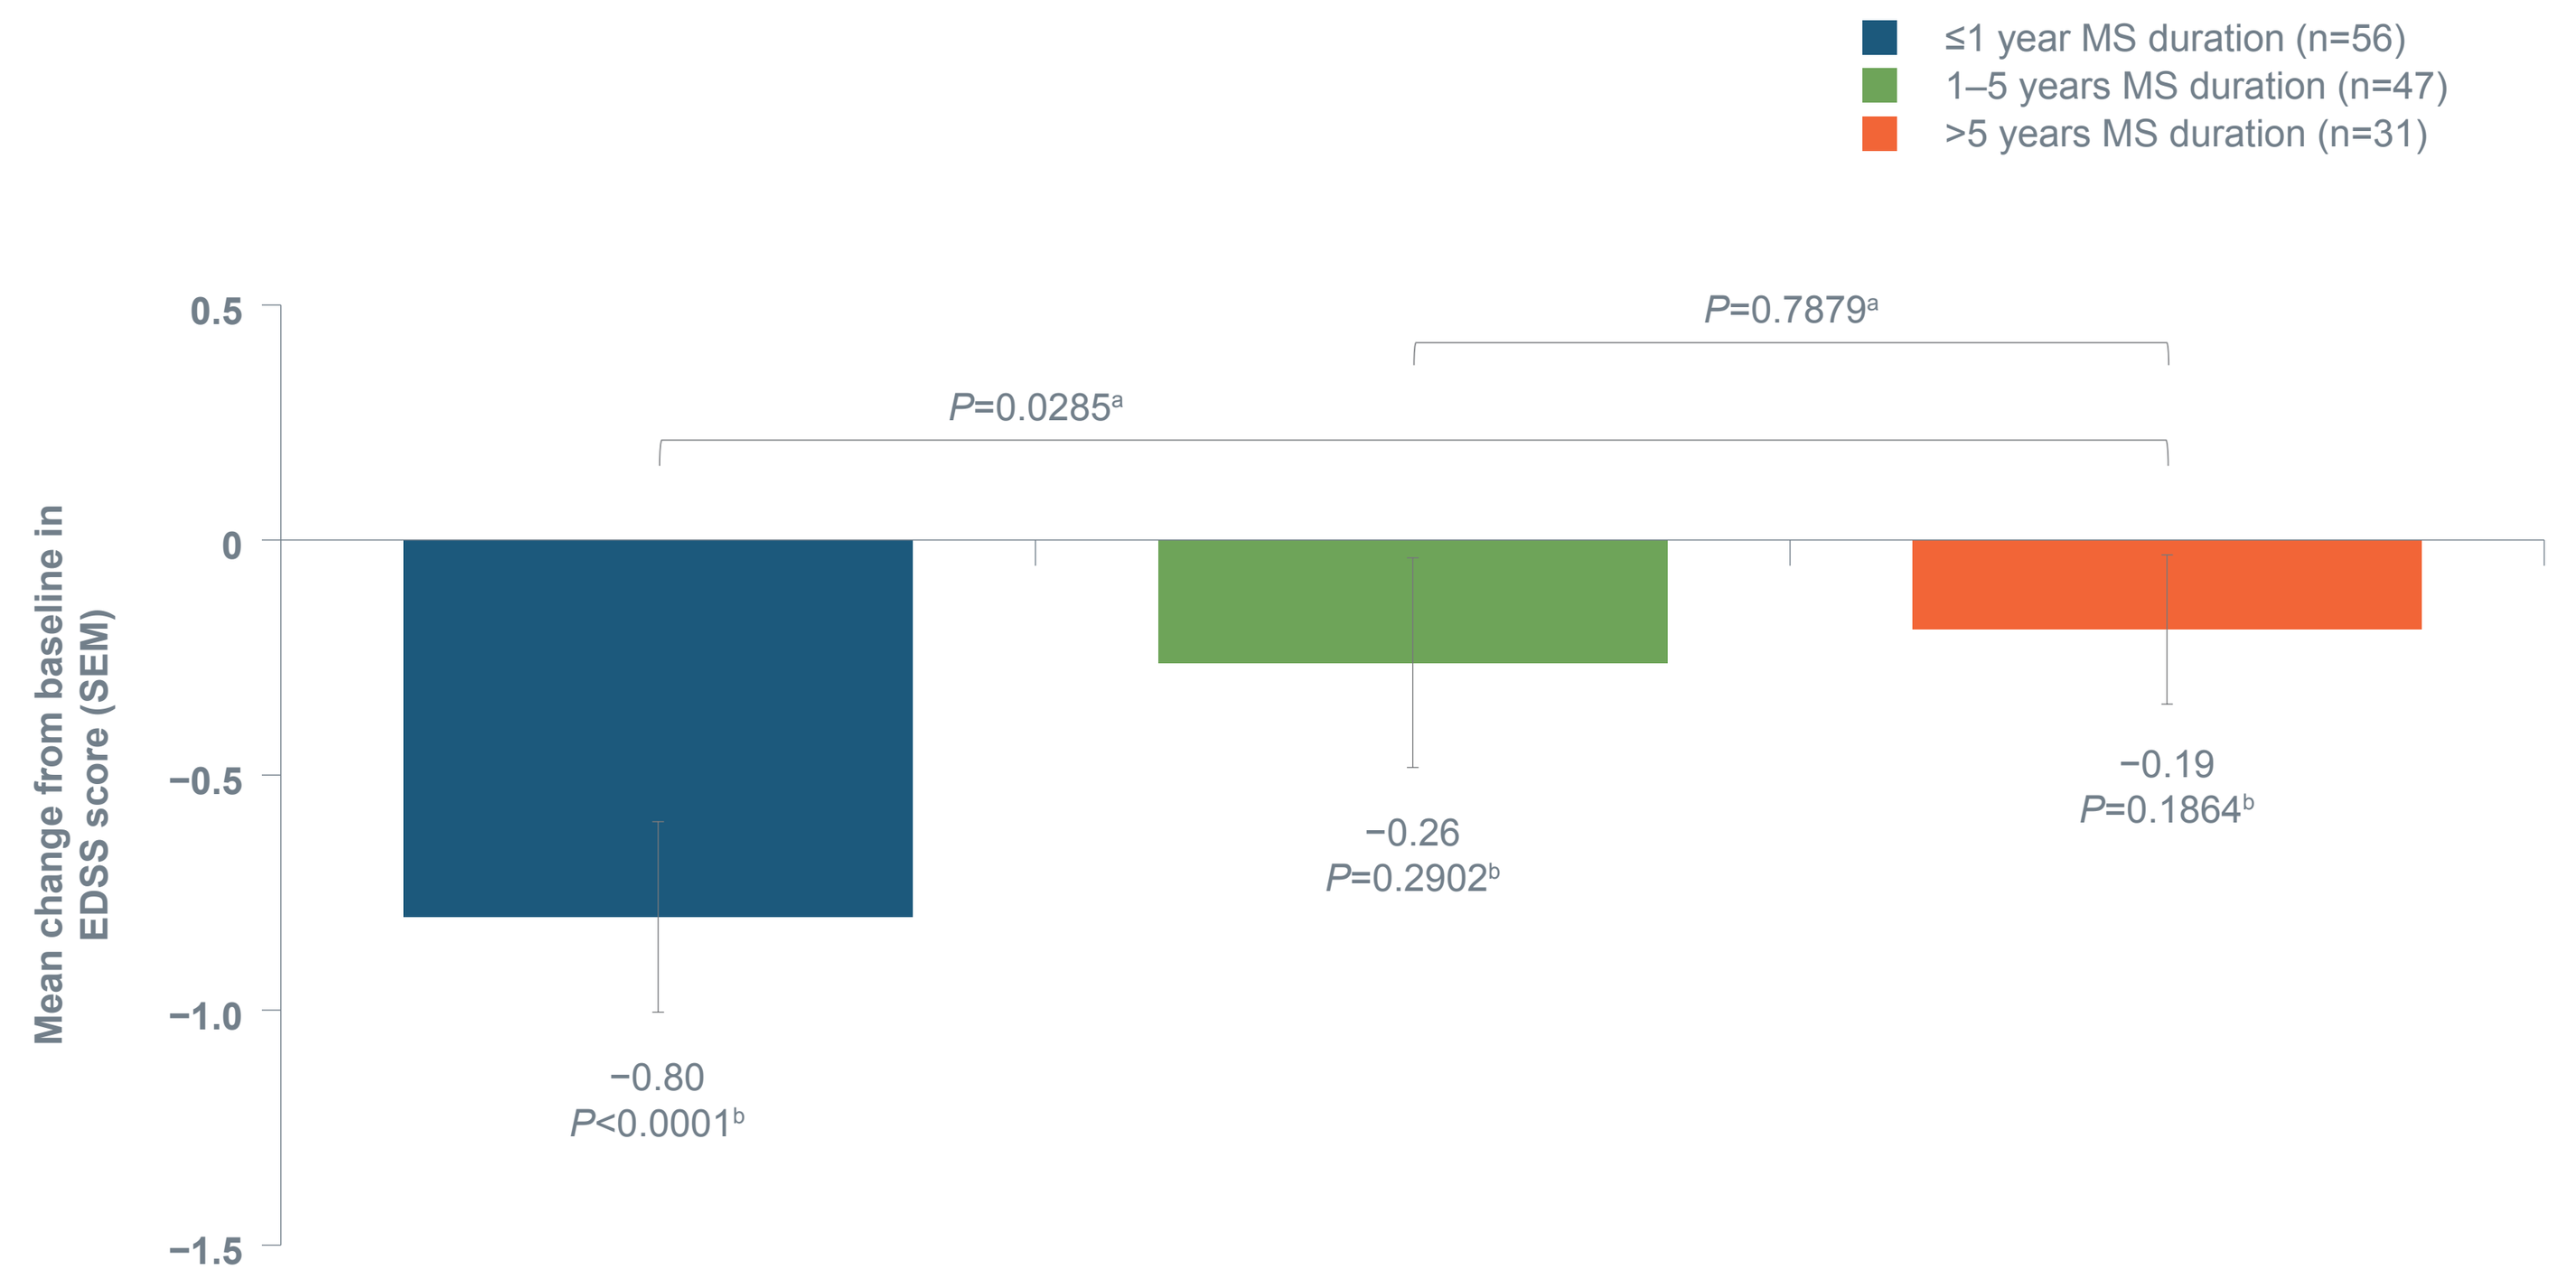
≤1 year MS duration (n=56)
1–5 years MS duration (n=47)
>5 years MS duration (n=31)
Mean change from baseline in EDSS score (SEM)
P=0.0285a
P=0.7879a
−0.80 P<0.0001b
−0.26 P=0.2902b
−0.19 P=0.1864b

SEM=standard error of the mean.
[a]Based on Wilcoxon rank-sum test between groups.
[b]Based on Wilcoxon signed-rank test within group.

Figure 3. Cumulative probability of 24-week confirmed EDSS worsening over 3 years of natalizumab treatment

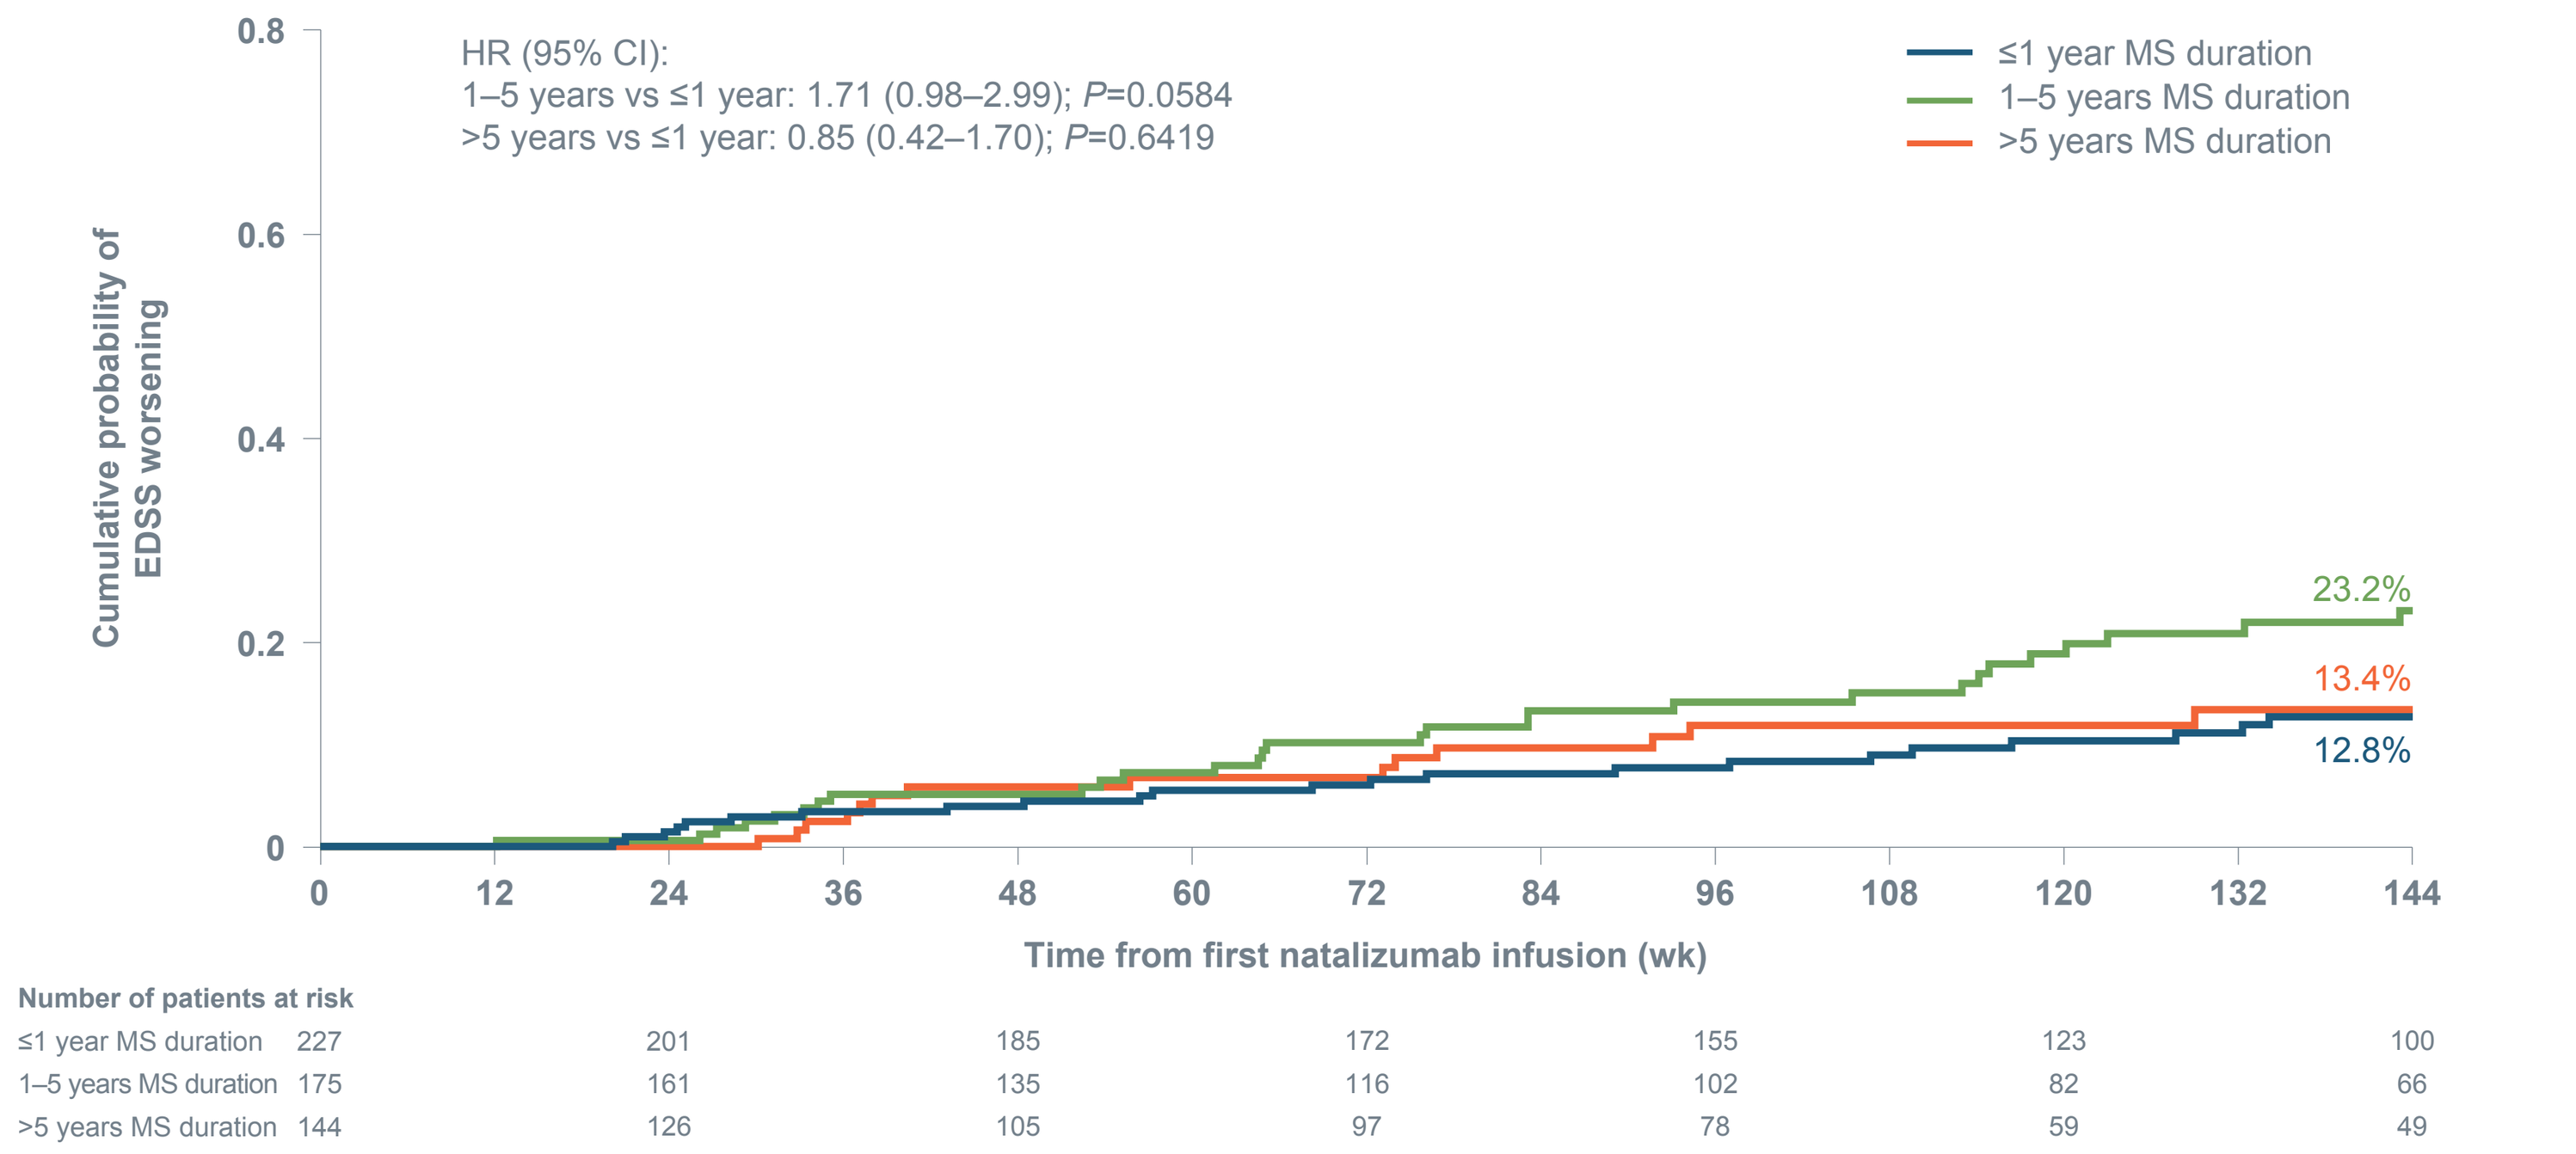
HR (95% CI):
1–5 years vs ≤1 year: 1.71 (0.98–2.99); P=0.0584
>5 years vs ≤1 year: 0.85 (0.42–1.70); P=0.6419
≤1 year MS duration
1–5 years MS duration
>5 years MS duration
Cumulative probability of EDSS worsening
23.2%
13.4%
12.8%
Time from first natalizumab infusion (wk)
Number of patients at risk
≤1 year MS duration 227 201 185 172 155 123 100
1–5 years MS duration 175 161 135 116 102 82 66
>5 years MS duration 144 126 105 97 78 59 49

HR=hazard ratio.
HRs and *P* values are based on a Cox model with age, number of relapses in the year prior to natalizumab initiation, and baseline EDSS category (EDSS score <3.0 or ≥3.0) as covariates. Data are recensored at week 144. Patients with available baseline EDSS scores are included.

Figure 4. Cumulative probability of 24-week confirmed EDSS improvement over 3 years of natalizumab treatment

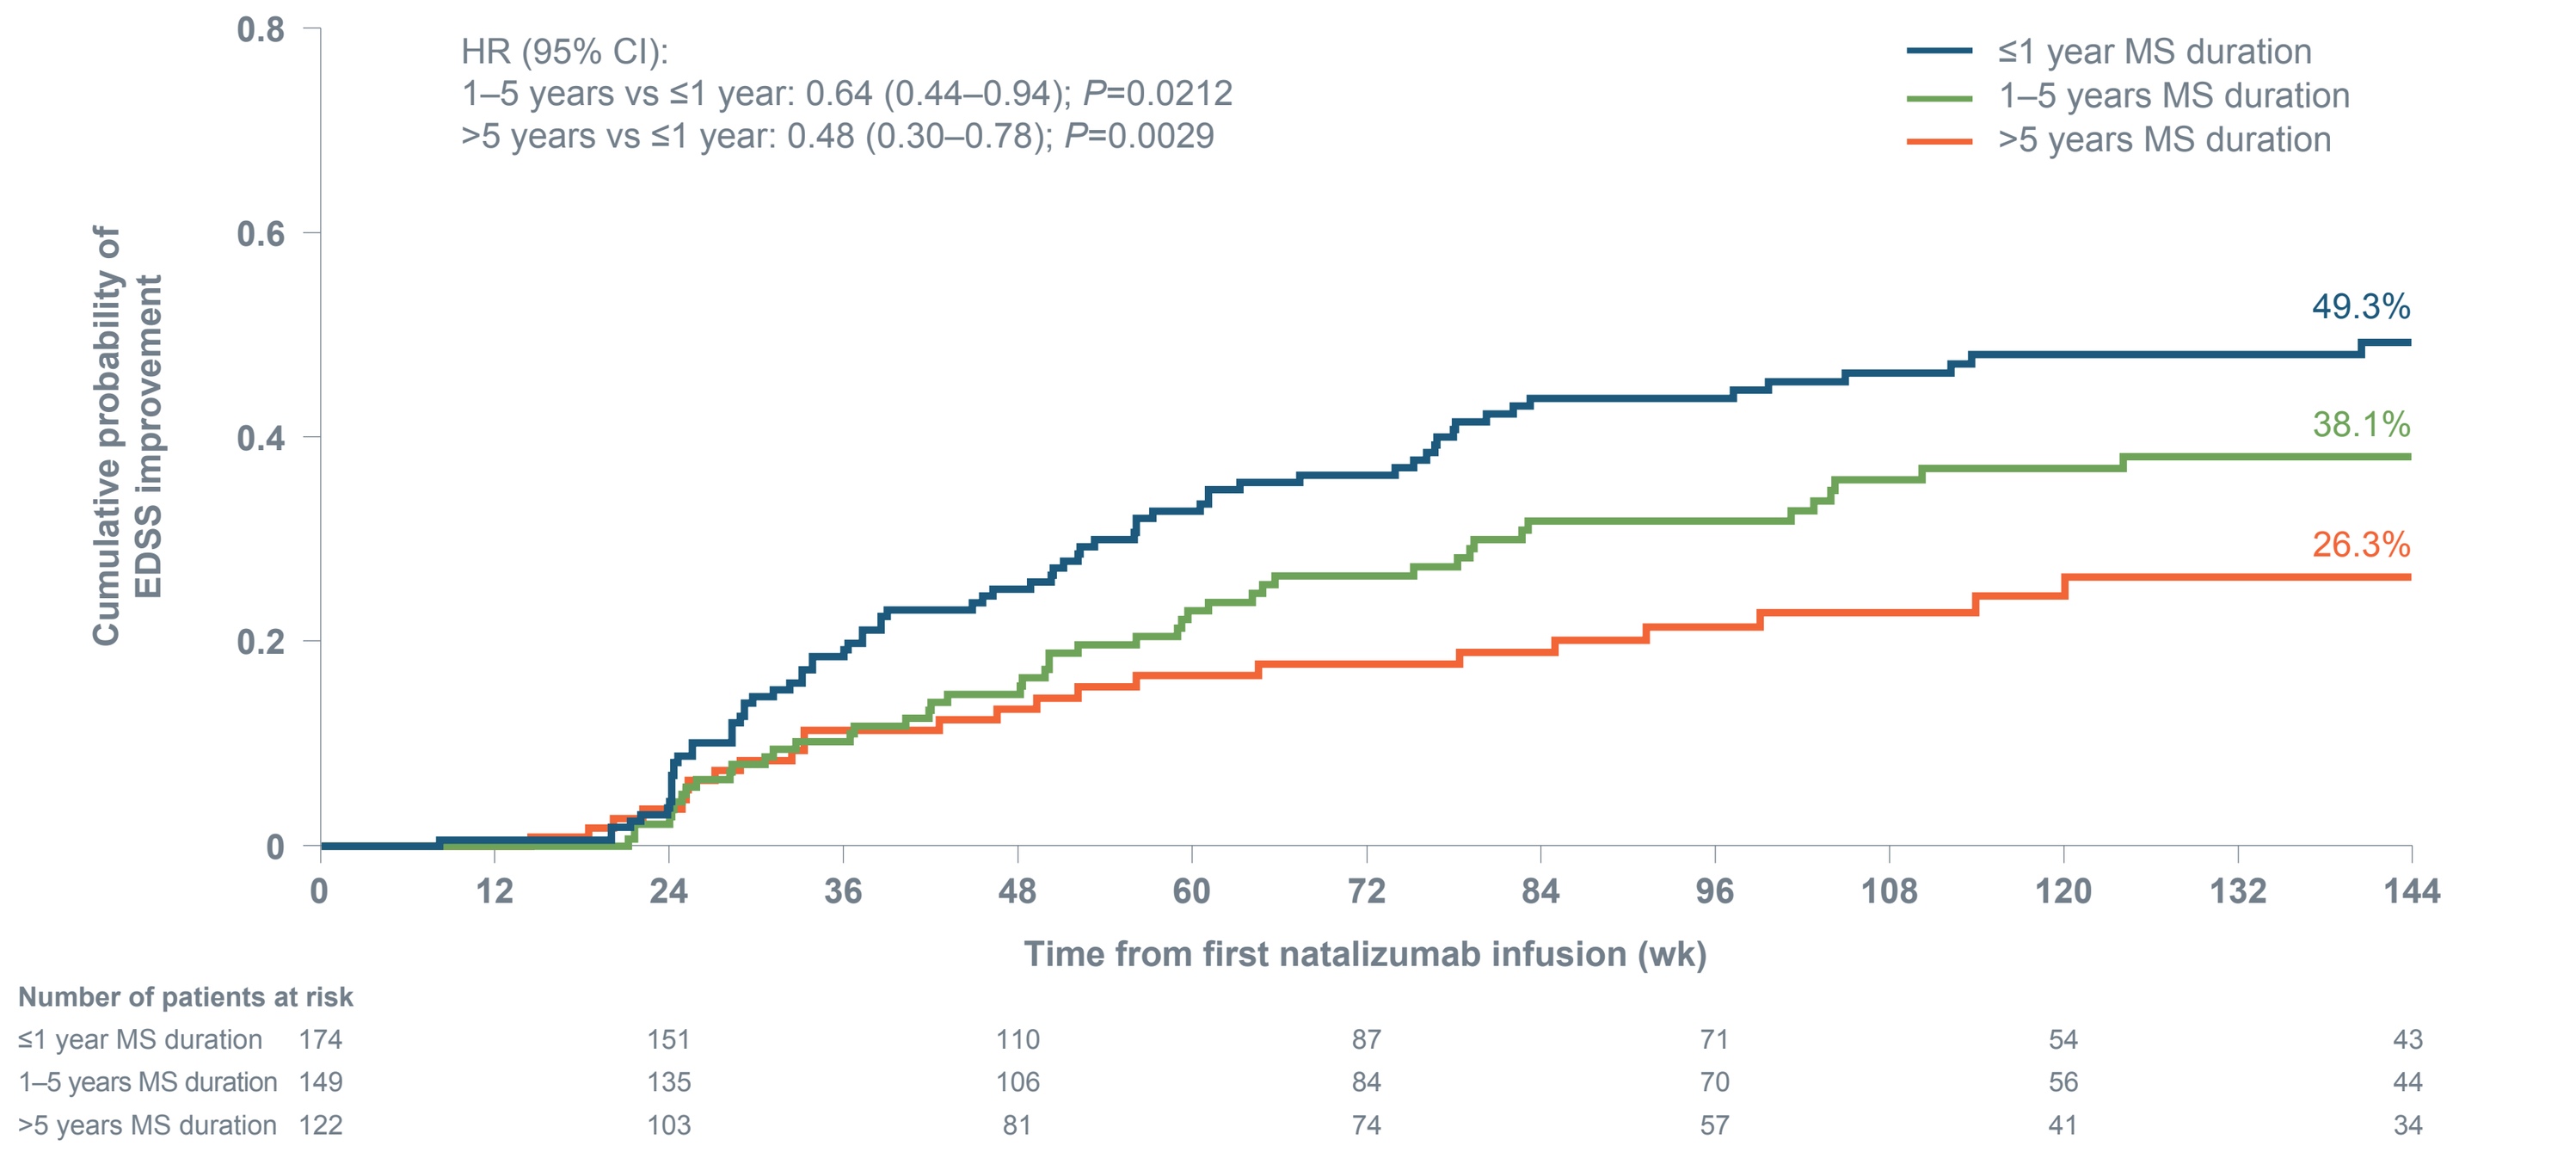
HR (95% CI):
1–5 years vs ≤1 year: 0.64 (0.44–0.94); P=0.0212
>5 years vs ≤1 year: 0.48 (0.30–0.78); P=0.0029
≤1 year MS duration
1–5 years MS duration
>5 years MS duration
Cumulative probability of EDSS improvement
49.3%
38.1%
26.3%
Time from first natalizumab infusion (wk)
Number of patients at risk
≤1 year MS duration 174 151 110 87 71 54 43
1–5 years MS duration 149 135 106 84 70 56 44
>5 years MS duration 122 103 81 74 57 41 34

HRs and *P* values are based on a Cox model with age, number of relapses in the year prior to natalizumab initiation, and baseline EDSS category (EDSS score <3.0 or ≥3.0) as covariates. Data are recensored at week 144. Patients with available baseline EDSS scores are included.

Table 1. Characteristics of treatment-naive patients in TOP (N=556)

Characteristic	MS duration ≤1 year (n=233)	MS duration 1–5 years (n=179)	MS duration >5 years (n=144)
Age, mean (SD), y[a]	**31.9 (9.96)**	**34.3 (10.08)**	**40.1 (9.19)**
Sex, female, n (%)	**157 (67.4)**	**119 (66.5)**	**103 (71.5)**
MS disease duration, y[a]			
Mean (SD)	0.5 (0.24)	2.4 (1.11)	12.4 (6.54)
Median (min, max)	0.5 (0, 1.0)	2.1 (1.0, 4.9)	10.7 (5.0, 42.7)
EDSS score[a]			
Mean (SD)	2.9 (1.58)	3.3 (1.60)	3.4 (1.65)
Median (min, max)	2.5 (0.0, 9.5)	3.0 (0.0, 7.0)	3.5 (0.0, 7.0)
Number of relapses in year prior[a]			
Mean (SD)	2.1 (0.93)	2.2 (1.03)	1.8 (0.93)
Median (min, max)	2 (0, 5)	2 (1, 7)	2 (0, 5)
Patients with Gd+ lesions, n (%)[a]	**144 (82.3)[b]**	**96 (80.0)[b]**	**63 (68.5)[b]**
Time on natalizumab, y			
Mean (SD)	3.1 (1.94)	3.1 (1.90)	2.7 (1.81)
Median (min, max)	2.9 (0, 7.9)	2.8 (0.1, 8.4)	2.3 (0, 8.4)

SD=standard deviation.
[a]Assessed at baseline.
[b]Data were available from 175 patients with MS duration ≤1 year, 120 patients with MS duration of 1–5 years, and 92 patients with MS duration >5 years.

References 1. Butzkueven H, *et al*. Presented at the American Academy of Neurology annual meeting; April 15–21, 2016; Vancouver, Canada. P2.069; 2. Kappos L, *et al*. *Mult Scler*. 2016;22(suppl 3):645-646. P1228; 3. Butzkueven H, *et al*. *J Neurol Neurosurg Psychiatry*. 2014;85:1190-1197; 4. Polman CH, *et al*. *N Engl J Med*. 2006;354:899-910; 5. Havrdova E, *et al*. *Mult Scler*. 2013;19(suppl 19):209-210. P519; 6. Phillips JT, *et al*. *Mult Scler*. 2011;17:970-979; 7. Giovannoni G, *et al*. *Mult Scler*. 2016;22(suppl 3):671-672. P1270; 8. Cadavid D, *et al*. *PLoS One*. 2013;8:e53297; 9. Prosperini L, *et al*. *J Neurol Neurosurg Psychiatry*. 2015;86:236-238; 10. Belachew S, *et al*. *Eur J Neurol*. 2011;18:240-245. **Disclosures** This study was funded by Biogen. TS: honoraria for consultancy and funding for travel from Biogen, Novartis. HB: compensation for consulting from Biogen, Merck Serono, Novartis; research support from Biogen, Merck Serono. LK: in the last 3 years, Institution (University Hospital Basel) received/used exclusively for research support: steering committee, advisory board, and consultancy fees from Actelion, Addex, Bayer HealthCare, Biogen, Biotica, Genzyme, Lilly, Merck, Mitsubishi, Novartis, Ono Pharma, Pfizer, Receptos, Sanofi, Santhera, Siemens, Teva, UCB, Xenoport; speaker fees from Bayer HealthCare, Biogen, Merck, Novartis, Sanofi, Teva; support of educational activities from Bayer HealthCare, Biogen, CSL Behring, Genzyme, Merck, Novartis, Sanofi, Teva; grants from Bayer HealthCare, Biogen, F. Hoffmann-La Roche, Merck, Novartis, the Swiss Multiple Sclerosis Society, the Swiss National Research Foundation, the European Union, the Roche Research Foundations. MT: scientific advisory boards for Biogen, Merck Serono, Novartis; speaker honoraria from Biogen, Merck Serono, Novartis, Sanofi, Teva; research grants from Biogen, Merck Serono, Novartis. HW: honoraria from Bayer, Biogen, Medac, Merck Serono, Novo Nordisk, Sanofi, Schering, Teva; consultant for Bayer Vital/Schering, Biogen, Medac, Merck Serono, Novartis, Novo Nordisk, Sanofi, Teva; research support from Bayer, Biogen, Medac, Merck Serono, Novo Nordisk, Sanofi, Schering, Teva. YC: employee of Biogen at the time this analysis was performed. NC, SL, PRH: employees of and own stock and/or stock options in Biogen. **Acknowledgments** All named authors meet the International Committee of Medical Journal Editors criteria for authorship for this poster and take responsibility for the integrity of the work as a whole. Biogen provided funding for medical writing support in the development of this poster; Alison Adams, PhD, of Ashfield Healthcare Communications (Middletown, CT) wrote the first draft of the poster based on input from authors, and Joshua Safran of Ashfield Healthcare Communications copyedited and styled the poster per congress requirements. Biogen reviewed and provided feedback on the poster to the authors. The authors had full editorial control of the poster and provided their final approval of all content.

American Academy of Neurology 2017 - 69th Annual Meeting | April 22-28, 2017 | Boston, MA