Real-world Data from over 10 Years in the TYSABRI® Observational Program: Long-term Safety and Effectiveness of Natalizumab in Relapsing-Remitting Multiple Sclerosis Patients

P908

Kappos L,[1] Butzkueven H,[2] Spelman T,[3] Trojano M,[4] Wiendl H,[5] Jiang X,[6] Kasliwal R,[6] Campbell N,[6] Ho P-R,[6] Licata S,[6] on behalf of the TYSABRI Observational Program (TOP) Investigators

[1]Neurologic Clinic and Policlinic, Departments of Medicine, Clinical Research, Biomedicine and Biomedical Engineering, University Hospital and University of Basel, Basel, Switzerland; [2]Department of Neuroscience, Central Clinical School, Alfred Campus, Monash University, Melbourne, Victoria, Australia, and Department of Neurology, Box Hill Hospital, Monash University, Box Hill, Victoria, Australia; [3]Department of Medicine and Melbourne Brain Centre at the Royal Melbourne Hospital, University of Melbourne, Melbourne, Victoria, Australia; [4]Department of Basic Medical Science, Neuroscience and Sense Organs, University of Bari, Bari, Italy; [5]Department of Neurology, University of Münster, Münster, Germany; [6]Biogen, Cambridge, MA, USA

Conclusions

- The TYSABRI Observational Program (TOP) began more than 10 years ago and is the largest ongoing real-world study of relapsing-remitting multiple sclerosis (RRMS) patients treated with natalizumab.
- This interim analysis of TOP extends the previous analysis[1] with a larger population (6148 vs 4821 patients), a longer natalizumab exposure duration (median of 38 vs 22 doses), and a longer follow-up time (median of 62 vs 26 months) to demonstrate natalizumab's consistent safety and effectiveness over 10 years.
- The effectiveness results confirm and broaden the findings from clinical trials.[2]
 - Significant annualised relapse rate (ARR) reductions were observed regardless of baseline disease activity or the number of prior treatments (with on-treatment reductions of 89.8%–94.3% from the year prior to natalizumab initiation).
 - Natalizumab provided long-term control of RRMS disease activity, as shown by the stability of relapse rates and Expanded Disability Status Scale (EDSS) scores over the study duration.
 - At 10 years, the estimated cumulative probability of disability improvement exceeded 30%, while the estimated probability of disability worsening remained <30%.
- For up to 10 years of natalizumab treatment, safety findings were consistent with the current safety profile of natalizumab,[3] and no new safety concerns were identified.
 - The incidence rates of opportunistic infections (OIs), progressive multifocal leukoencephalopathy (PML), and malignancy were low (0.002–0.003 events per patient-year).
- Long-term, real-world studies like TOP provide valuable information for clinicians and patients considering the benefit-risk profile of RRMS therapies.
- The planned continued follow-up for up to 15 years in TOP will provide the longest evaluation of real-world outcomes in natalizumab-treated patients.

Introduction

- Natalizumab is a highly efficacious therapy for the treatment of RRMS, as demonstrated in clinical trials and a number of real-world studies.[2,4]
- TOP, which began >10 years ago, is the largest ongoing real-world study of natalizumab-treated patients with RRMS.
 - TOP collects data on safety and clinical outcomes in patients with RRMS treated with natalizumab in clinical practice.[1]
 - The 5-year interim analysis of TOP did not reveal new safety signals associated with natalizumab treatment. The overall relapse rate was significantly reduced from prenatalizumab levels and remained low, and disability levels remained stable over time.[1] These results supported the efficacy demonstrated in the pivotal trials[2] with real-world data.
- The current analysis reports on 10-year interim data on the long-term safety and effectiveness of natalizumab in TOP patients treated in real-world settings.

Objectives

- To report an interim analysis of safety and effectiveness in patients with up to 10 years of natalizumab treatment in TOP.

Methods

- TOP is an ongoing, open-label, multinational, multicentre prospective observational study of patients with RRMS initiating natalizumab in clinical practice settings (ClinicalTrials.gov, NCT00493298).[1]
- This interim analysis includes data from study initiation (July 2007) through November 2017.
- ARRs for the year prior to starting natalizumab and the period during which patients were on natalizumab (and ≤84 days after the last dose) were compared using a repeated Poisson model.
 - ARRs were also evaluated by baseline characteristics: median age, number of relapses in the prior year, EDSS score category, and number and type of prior disease-modifying therapies (DMTs) used.
- The cumulative probabilities of 24-week–confirmed disability worsening (EDSS score increase of ≥1.5 from a baseline score of 0.0, ≥1.0 from a score of 1.0–5.5, or ≥0.5 from a score ≥6.0) and 24-week–confirmed disability improvement (decrease of ≥1.0 from a baseline score ≥2.0) were estimated using the Kaplan-Meier method. Confirmation could occur ≤84 days after the last dose of natalizumab.
- Serious adverse events (SAEs) were assessed at clinical visits beginning with the first dose of natalizumab. SAEs that occurred up to 6 months after the last natalizumab infusion were included, except for PML and death, which were included after the first dose irrespective of the date of occurrence.
- The incidence rates of SAEs of interest (OIs, malignancy, and PML) were examined overall and in the first 3 years and beyond 3 years on treatment (reflecting median exposure) to determine if incidence changed over time.
 - Incidence rate per patient-year was calculated as the number of patients with an event divided by the number of patient-years of follow-up overall or during the first 3 years or beyond 3 years on treatment. Exact confidence intervals (CIs) were calculated based on the Poisson distribution.

Results

Patient enrolment and disposition

- As of 1 November 2017, 6148 patients were enrolled in TOP (Figure 1). Baseline characteristics are shown in Table 1.
- A total of 3210 patients (52.2%) discontinued natalizumab but remained in TOP; 2117 (34.4%) withdrew from TOP.
 - The most common reason reported for discontinuation was positive anti–JC virus (JCV) antibody serostatus (n=1487; 24.2%).
- Median exposure was 38 (range, 1–135) doses; median follow-up time was 62 (range, 1–130) months.

Relapses

- The ARR decreased from 1.99 in the year prior to starting natalizumab to 0.24 during the first year of natalizumab treatment and 0.19 during the second year; yearly ARRs remained <0.20 in years 3–10 (Figure 2). The overall ARR while patients were on natalizumab was 0.15 (95% CI: 0.14–0.15), a 92.5% reduction from the year prior to starting natalizumab.
 - When patients were stratified by baseline characteristics, the on-treatment ARR reduction from the year prior to initiation ranged from 89.8% to 94.3% (Figure 3).
 - The greatest reductions in ARR (>94.0%) were observed among patients with the lowest baseline EDSS scores (≤1.5) and those who were treatment naive at natalizumab initiation (Figure 3C–E).
- The majority of patients (n=3878; 63%) were relapse free, and the estimated cumulative probability of remaining relapse free over 10 years was 45.8%.

Disability

- In TOP, 810 of 6148 (13.2%) patients had confirmed disability worsening, and 1210 of 5179 (23.4%) had confirmed disability improvement.
- At 10 years, the estimated cumulative probabilities of 24-week confirmed disability worsening and improvement were 27.8% and 33.1%, respectively, though numbers of patients at risk after 9 years were low. At 9 years, probabilities were 25.0% and 32.3%, respectively (Figure 4).

Safety

- Overall, 13.5% of patients experienced ≥1 SAE; 4.7% had ≥1 SAE reported to be treatment related. There were 30 deaths (0.5%).
- The most commonly reported SAEs by Medical Dictionary for Regulatory Activities (MedDRA) preferred term were immune reconstitution inflammatory syndrome (IRIS) and PML (Table 2).
- Incidence rates for OI, malignancy, and PML were low (Table 3) and were similar during the first 3 years on treatment and after 3 years, as indicated by overlapping CIs.
 - The most commonly reported OI was PML (n=52), and the most commonly reported malignancy was breast cancer (n=12).

Figure 1. Annual accounting of patients in TOP

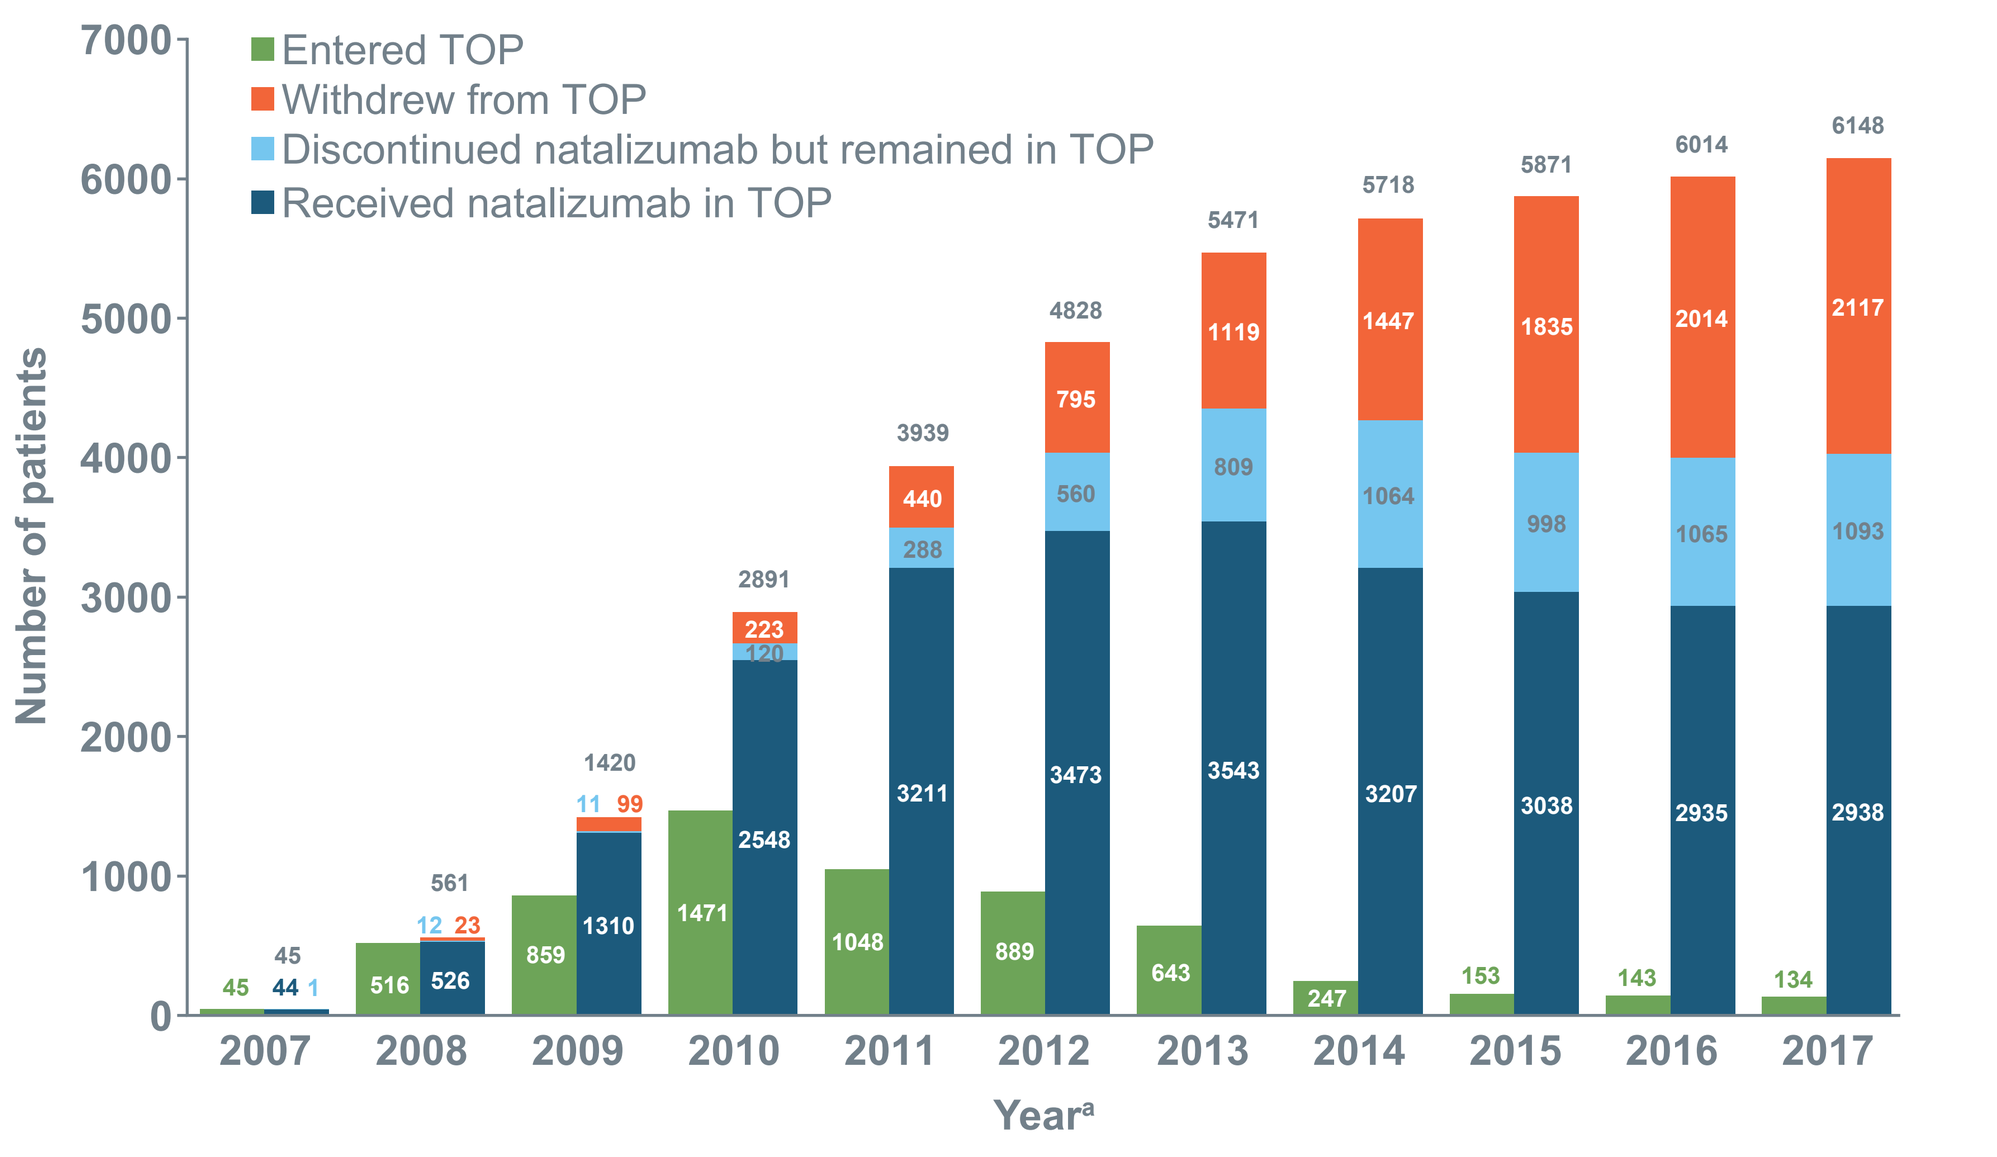
Entered TOP
Withdrew from TOP
Discontinued natalizumab but remained in TOP
Received natalizumab in TOP
Number of patients
Year[a]
2007 2008 2009 2010 2011 2012 2013 2014 2015 2016 2017
45 561 1420 2891 3939 4828 5471 5718 5871 6014 6148

Patients who received natalizumab in TOP are those who remained in TOP and continued natalizumab treatment through 1 November of the given year. Cumulative numbers of patients who withdrew over the study duration are shown.
[a]Accounting in 2007 is shown for study start (July 2007) through 1 November 2007. For each year, patients enrolled after 1 November are accounted for in the following year.

Figure 2. ARR during each year of natalizumab treatment

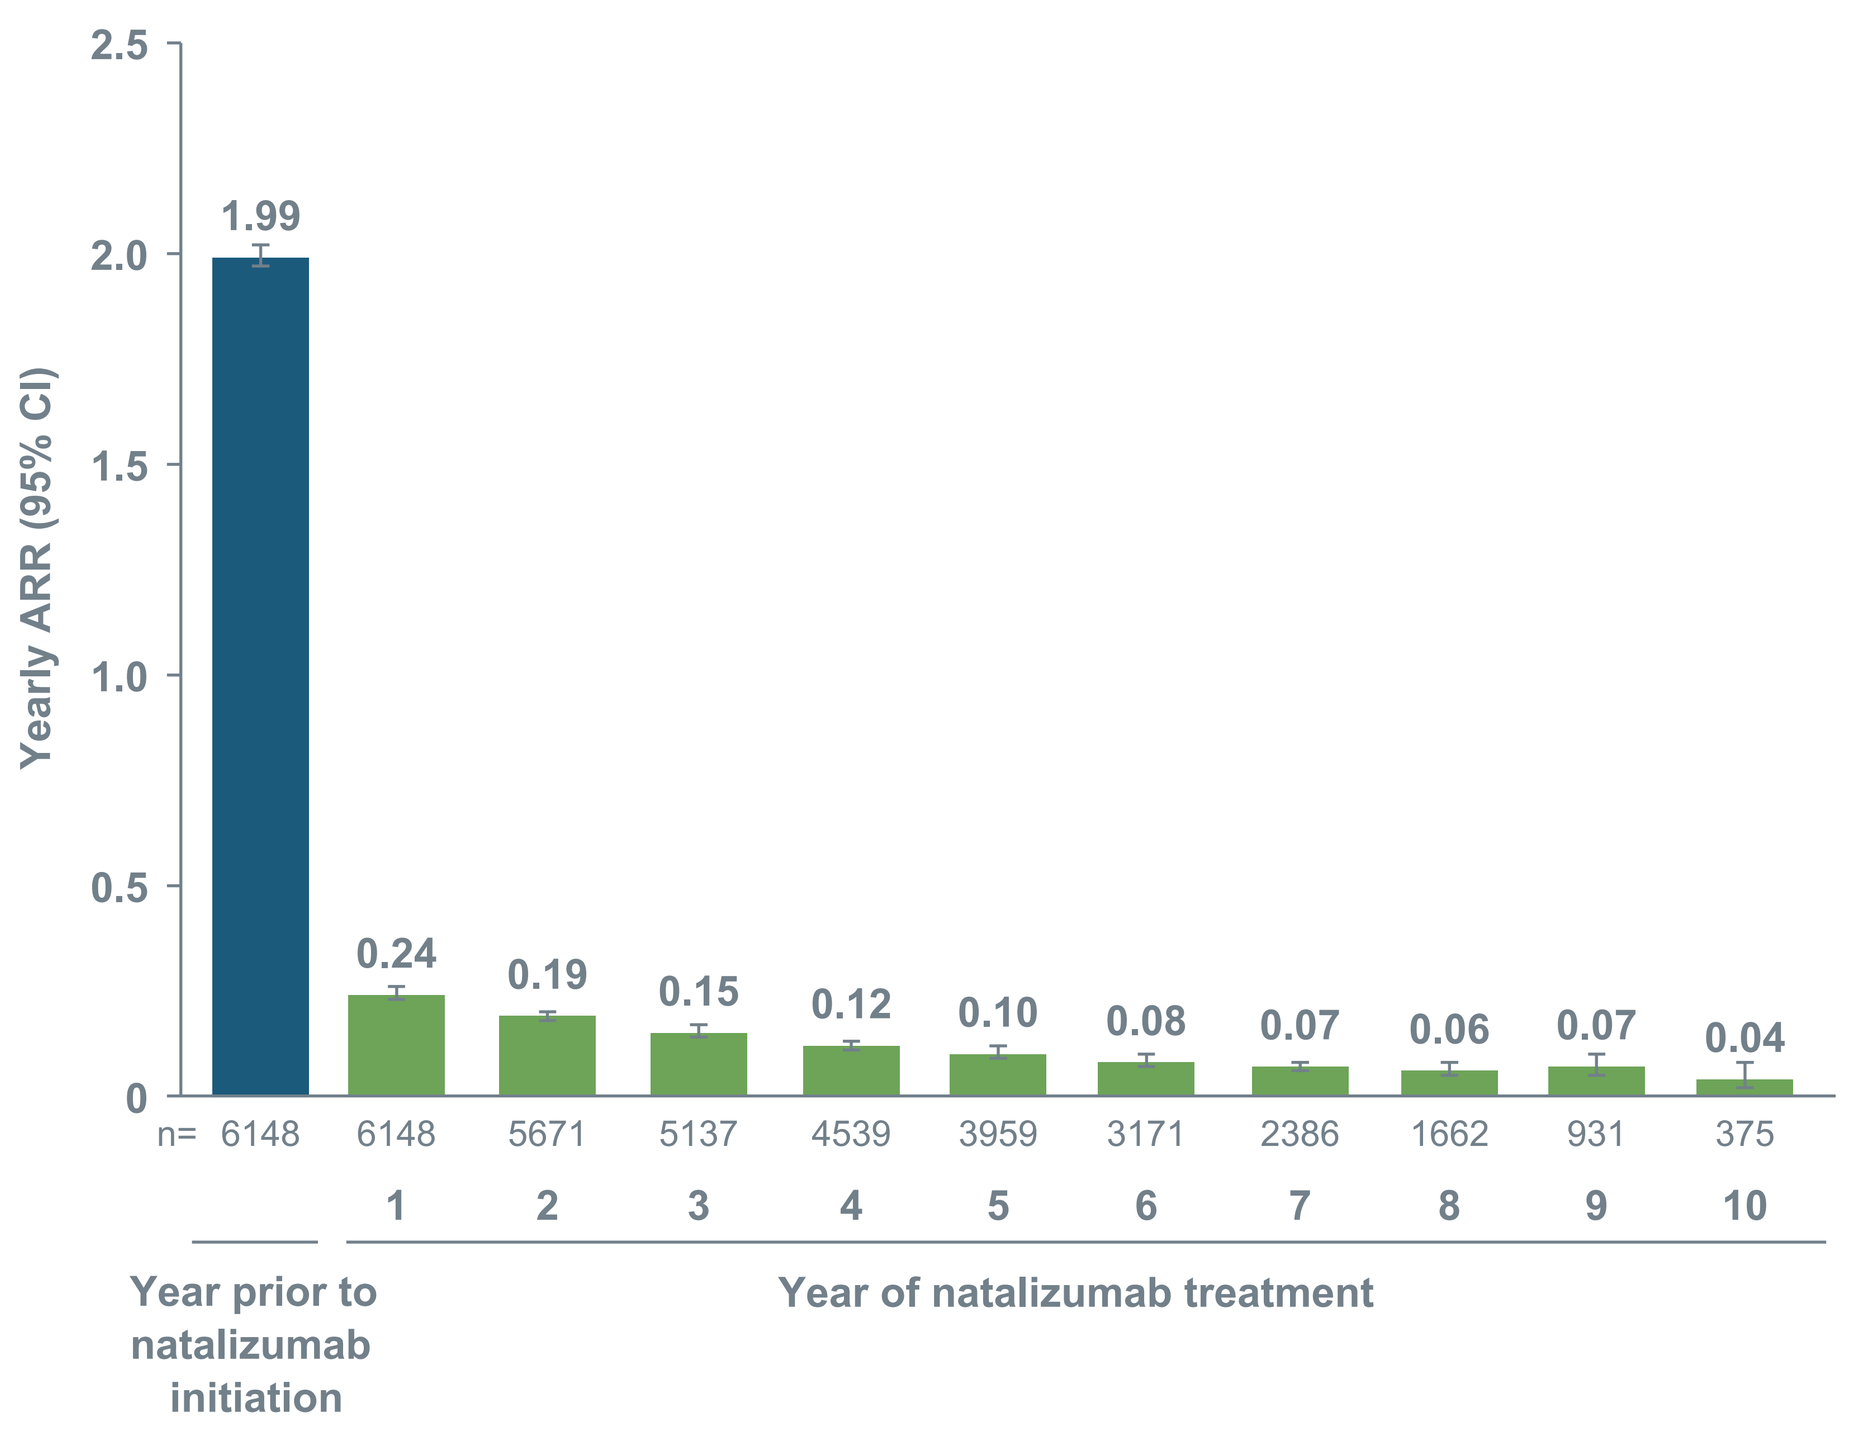
Yearly ARR (95% CI)
1.99
0.24 0.19 0.15 0.12 0.10 0.08 0.07 0.06 0.07 0.04
n= 6148 6148 5671 5137 4539 3959 3171 2386 1662 931 375
1 2 3 4 5 6 7 8 9 10
Year prior to natalizumab initiation
Year of natalizumab treatment

Figure 3. ARRs in the year prior to natalizumab treatment and after starting natalizumab by patient characteristics and treatment history at baseline

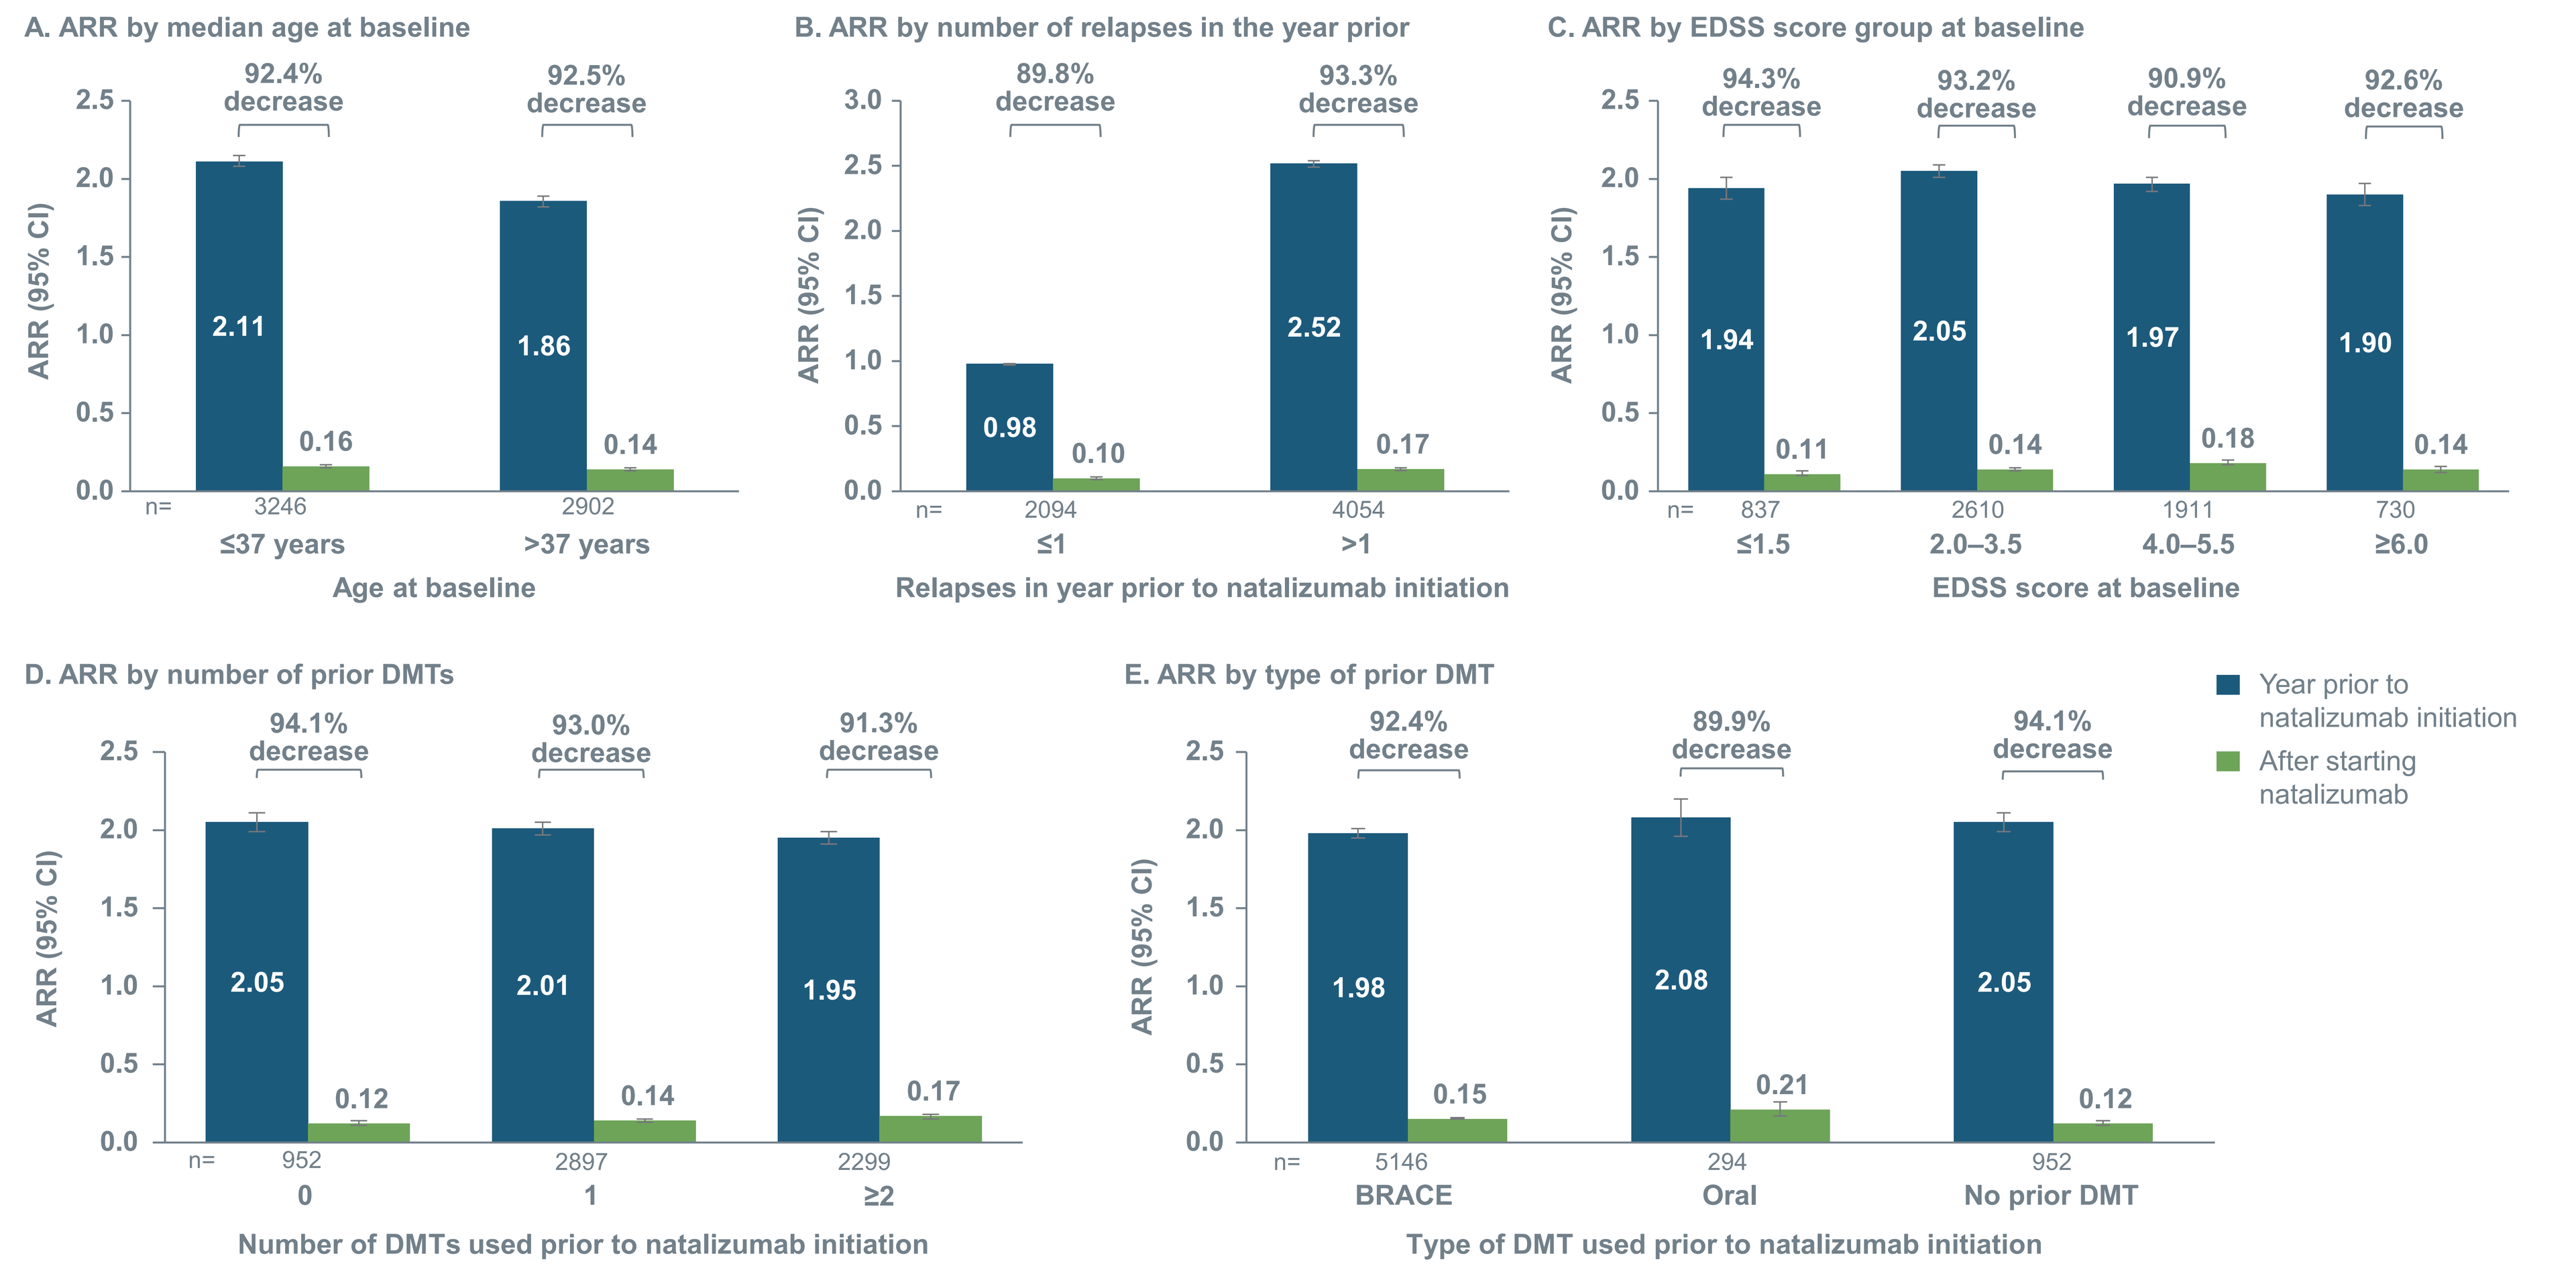
A. ARR by median age at baseline
92.4% decrease
92.5% decrease
2.11 0.16
1.86 0.14
n= 3246 2902
≤37 years >37 years
Age at baseline
B. ARR by number of relapses in the year prior
89.8% decrease
93.3% decrease
0.98 0.10
2.52 0.17
n= 2094 4054
≤1 >1
Relapses in year prior to natalizumab initiation
C. ARR by EDSS score group at baseline
94.3% decrease 93.2% decrease 90.9% decrease 92.6% decrease
1.94 0.11 2.05 0.14 1.97 0.18 1.90 0.14
n= 837 2610 1911 730
≤1.5 2.0–3.5 4.0–5.5 ≥6.0
EDSS score at baseline
D. ARR by number of prior DMTs
94.1% decrease 93.0% decrease 91.3% decrease
2.05 0.12 2.01 0.14 1.95 0.17
n= 952 2897 2299
0 1 ≥2
Number of DMTs used prior to natalizumab initiation
E. ARR by type of prior DMT
92.4% decrease 89.9% decrease 94.1% decrease
1.98 0.15 2.08 0.21 2.05 0.12
n= 5146 294 952
BRACE Oral No prior DMT
Type of DMT used prior to natalizumab initiation
Year prior to natalizumab initiation
After starting natalizumab
ARR (95% CI)

BRACE=Betaseron®, Rebif®, Avonex®, Copaxone®, Extavia®.

Figure 4. Estimated cumulative probabilities of (A) confirmed disability worsening and (B) confirmed disability improvement at 9 years of natalizumab treatment

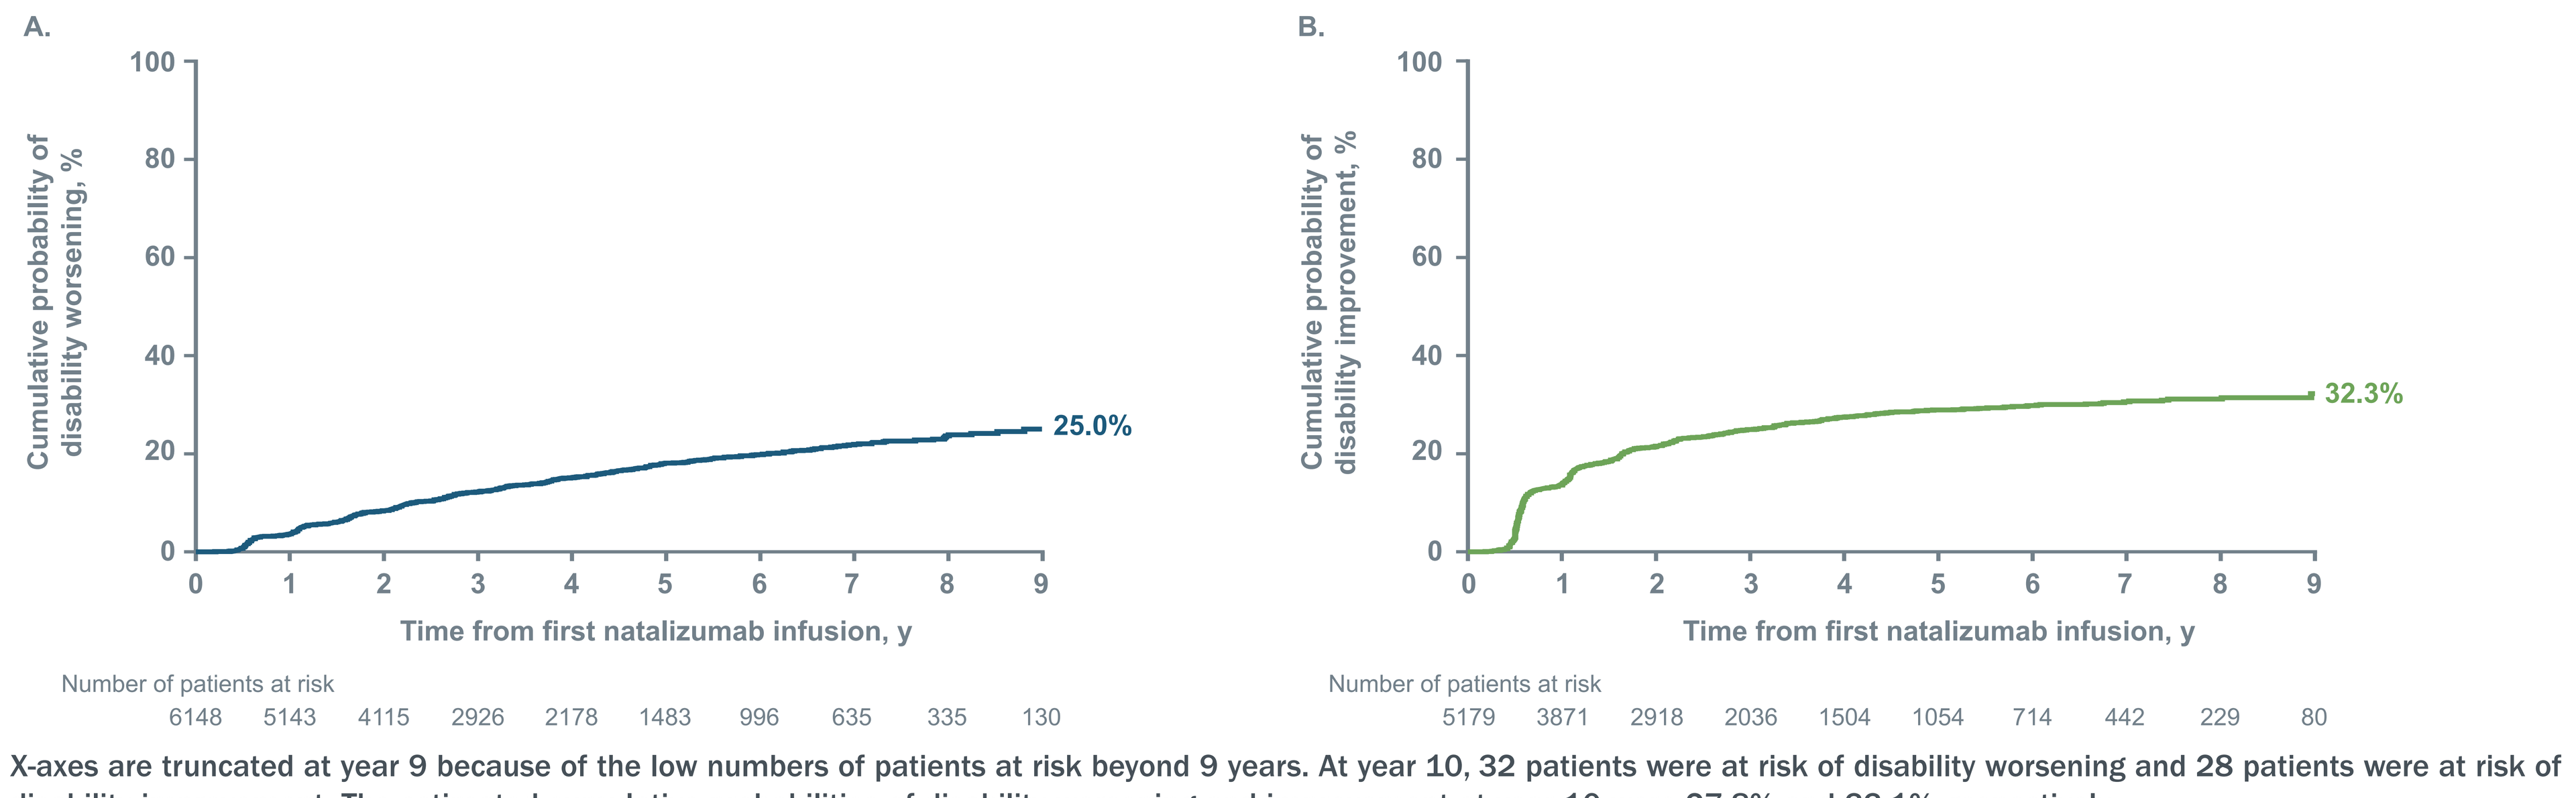
A.
Cumulative probability of disability worsening, %
25.0%
Time from first natalizumab infusion, y
Number of patients at risk
6148 5143 4115 2926 2178 1483 996 635 335 130
B.
Cumulative probability of disability improvement, %
32.3%
Time from first natalizumab infusion, y
Number of patients at risk
5179 3871 2918 2036 1504 1054 714 442 229 80

X-axes are truncated at year 9 because of the low numbers of patients at risk beyond 9 years. At year 10, 32 patients were at risk of disability worsening and 28 patients were at risk of disability improvement. The estimated cumulative probabilities of disability worsening and improvement at year 10 were 27.8% and 33.1%, respectively.

Table 1. Baseline demographics and clinical characteristics

Baseline characteristic	N=6148
Age, mean (SD), y	37.1 (9.77)
Female, n (%)	4430 (72.1)
Disease duration at baseline, median (range), y	7.8 (0–48)
Relapses in year prior to natalizumab initiation, mean (SD)	2.0 (1.00)
Baseline EDSS score, mean (SD)	3.5 (1.62)
DMT duration prior to natalizumab initiation, y	
Mean (SD)	4.0 (3.85)
Median (range)	2.9 (0–22)
Prior DMTs, n (%)	
0	952 (15.5)
1	2897 (47.1)
≥2	2299 (37.4)
Patients who were anti-JCV antibody positive, n (%)	2102 (34.2)[a]

[a]Data on anti-JCV antibody status at enrolment were available for 4339 patients.
SD=standard deviation.

Table 2. Incidence of SAEs occurring in ≥10 patients in TOP

MedDRA preferred term, n (%)[a]	N=6148
IRIS	54 (0.9)
PML, confirmed	52 (0.8)
Abortion spontaneous	37 (0.6)
Hypersensitivity	26 (0.4)
Pneumonia	23 (0.4)
Urinary tract infection	20 (0.3)
Fall	19 (0.3)
Depression	18 (0.3)
Epilepsy	18 (0.3)
Herpes zoster	17 (0.3)
Breast cancer	12 (0.2)
Intervertebral disc protrusion	12 (0.2)
Suicide attempt	11 (0.2)
Escherichia urinary tract infection	10 (0.2)
Pulmonary embolism	10 (0.2)
Pyelonephritis	10 (0.2)

[a]Each patient was counted only once within each preferred term. Multiple sclerosis was also reported as an SAE in 15 patients.

Table 3. Incidence of opportunistic infection, malignancy, and PML

	Overall exposure (26,060 patient-years; N=6148)		First 3 years on treatment (15,773 patient-years; N=6148)		Beyond 3 years on treatment (10,245 patient-years; N=3719)	
SAE type	Patients with event, n (%)	Incidence rate (95% CI), events per patient-year	Patients with event, n (%)	Incidence rate (95% CI), events per patient-year	Patients with event, n (%)	Incidence rate (95% CI), events per patient-year
OI	71 (1.2)	0.003 (0.002–0.003)	28 (0.5)	0.002 (0.001–0.003)	44 (1.2)	0.004 (0.003–0.006)
Malignancy	59 (1.0)	0.002 (0.002–0.003)	33 (0.5)	0.002 (0.001–0.003)	26 (0.7)	0.003 (0.002–0.004)
PML, confirmed	52 (0.8)	0.002 (0.002–0.003)	17 (0.3)	0.001 (0.001–0.002)	35 (0.9)	0.003 (0.002–0.005)

References 1. Butzkueven H, *et al. J Neurol Neurosurg Psych*. 2014;85:1190-1197; 2. Polman CH, *et al. N Engl J Med*. 2006;354:899-910; 3. TYSABRI (natalizumab) [summary of product characteristics]. Maidenhead, UK: Biogen; 2018; 4. Van Pesch V, *et al. Clin Neurol Neurosurg*. 2016;149:55-63.
Disclosures This study was supported by Biogen. LK: institution (University Hospital Basel) received in the last 3 years and used exclusively for research support at the department: steering committee, advisory board and consultancy fees from Actelion, Alkermes, Almirall, Bayer, Biogen, Celgene Receptos, df-mp, Excemed, GeNeuro, Genzyme, Japan Tobacco, Merck, Minoryx, Mitsubishi Pharma, Novartis, Roche, Sanofi, Santhera, Teva, Vianex; licensing fees for Neurostatus-UHB products. The research of the MS Centre in Basel has been supported by grants from Bayer, Biogen, Novartis, the Swiss MS Society, the Swiss National Research Foundation, the European Union, the Roche Research Foundations. HB: compensation for steering committee, advisory board, and consultancy fees from Biogen, Merck, Novartis, Oxford PharmaGenesis, Roche, Teva; research support from Biogen, Merck, NHMRC Australia, Novartis, MS Research Australia, UK MS Trust. TS: honoraria for consultancy and funding for travel from Biogen, Novartis. MT: compensation for consulting from Biogen, Merck Serono, Novartis; speaker honoraria from Biogen, Merck Serono, Novartis, Sanofi, Teva; research grants from Biogen, Merck Serono, Novartis. HW: honoraria from AbbVie, Actelion, Alexion, Biogen, Cognomed, Evgen Pharma, Hoffmann-La Roche, MedDay, Merck Serono, Novartis, Roche, Sanofi Genzyme, Teva; research support from Biogen, GlaxoSmithKline, Roche, Sanofi Genzyme. XJ, KR, NC, P-RH, SL: employees of and may hold stock and/or have stock options in Biogen.
Acknowledgements The authors gratefully acknowledge the contributions of the TOP study investigators. All named authors meet the International Committee of Medical Journal Editors criteria for authorship for this poster and take responsibility for the integrity of the work as a whole. Biogen provided funding for medical writing support in the development of this poster; Nicole Farra, PhD, and Alison Adams, PhD, of Ashfield Healthcare Communications (Middletown, CT, USA) wrote the first draft of the poster based on input from authors, and Shannon Davis and Joshua Safran of Ashfield Healthcare Communications copyedited and styled the poster per congress requirements. Biogen reviewed and provided feedback on the poster to the authors. The authors had full editorial control of the poster and provided their final approval of all content.

34th Congress of the European Committee for Treatment & Research in Multiple Sclerosis ECTRIMS (2018) | October 10–12, 2018 | Berlin, Germany

Biogen-05151 - Version January 2019